Phaxan$_{CD}$™, a Captisol®-enabled water soluble preparation of alphaxalone for intravenous anesthesia and sedation: comparison of toxicity with propofol and Althesin®

CS Goodchild[1,2], A Kolosov[1], B Boyd[3], and JM Serrao[2].
1 Monash Institute of Medical Research, 27-31 Wright Street, Clayton, 3168 Victoria, Australia
2 Goodchild Investments, Suite 200, 45 Glenferrie Rd, Malvern, 3144 Victoria, Australia
3 Faculty of Pharmacy and Pharmaceutical Sciences, Monash University (Parkville Campus), 381 Royal Parade, Parkville, 3052 Victoria, Australia

ABSTRACT

Introduction Alphaxalone is a neuroactive steroid anesthetic. This water-insoluble drug was formulated using CremophorEL (Althesin®)[1], found to be a safe (therapeutic index (LD50÷AD50) of 15-20)[2] and versatile intravenous anesthetic used in clinical practice in many countries from 1972-1984. [note the therapeutic index of propofol is 6][3]. Althesin® was withdrawn from clinical practice because of hypersensitivity to the CremophorEL. Previous attempts to make an aqueous formulation of alphaxalone suitable for human use have failed. Phaxan$_{CD}$™ is a solution of alphaxalone 10mg/ml dissolved in 0.9% saline and 13% Captisol® (7-sulfobutylether β-cyclodextrin; a molecule with a lipophilic cavity that enables drug dispersal in water for human use).

Objectives measure the doses of Phaxan$_{CD}$™ that cause anesthesia and lethality in rats and
compare the therapeutic index of Phaxan$_{CD}$™ with propofol and Althesin®

The studies below were approved by Monash University IRB.

Methods Alphaxalone (10mg.ml^{-1}) was prepared using 13% Captisol® and saline (Phaxan$_{CD}$® - PHAX). An "Althesin®-like" solution of alphaxalone was prepared in 20% CremophorEL (ALTH) as described previously[2]. Jugular intravenous catheters were implanted in male Wistar rats (150-200g). Separate groups of ten rats each were given intravenous injections of PHAX, ALTH or propofol emulsion [10mg.ml^{-1}; PROP] from 1.25 mg.kg^{-1} to lethal doses. Doses of each agent that caused anaesthesia (loss of righting reflex) and lethality in 50% of rats (AD_{50} and LD_{50}) were calculated by probit analysis.

Results Intravenous PHAX, ALTH and PROP caused dose-related sedation and anesthesia, AD_{50} values being 2.79, 2.95 and 4.63 mg.kg^{-1} respectively. Propofol was 100% lethal at doses greater than 30mg.kg^{-1} (LD_{50} = 30.3 mg/kg). At doses of alphaxalone between 50 and 60mg.kg^{-1} all rats given Althesin® died (LD_{50} = 43.6 mg/kg) whereas none died when given the same doses of alphaxalone as Phaxan$_{CD}$™ which caused no more than 20% lethality even at the maximum dose tested of 84 mg.kg^{-1}. Control experiments with the vehicles of the three preparations given alone showed no toxicity but the presence of Captisol® increased the therapeutic index of alphaxalone in PHAX compared with ALTH.

SEE TABLE 1

Conclusions Both alphaxalone formulations (Phaxan$_{CD}$™ and Althesin®) are equipotent anesthetics, causing fast onset anesthesia with twice the potency of propofol. PhaxanCD™ is less toxic than the propofol lipid formulation and also the alphaxalone formulation in CremophorEL (Althesin®).

TABLE 1

	Althesin®	Phaxan$_{CD}$	propofol
dose causing all 10 rats to lose righting reflex mg.kg^{-1}	5	5	10
AD_{50} for loss of righting reflex mg.kg^{-1}	2.95	2.79	4.63
AD_{95} for loss of righting reflex mg.kg^{-1}	4.39	4.26	5.40
AD_{50} for loss of tail pinch response reflex mg.kg^{-1}	6.46	6.56	8.40
AD_{95} for loss of tail pinch response mg.kg^{-1}	14.09	8.56	14.46
duration (minutes) of loss of righting reflex at dose causing all 10 rats to lose righting reflex mean (SD)	3.6 (2.18)	1.9 (0.84)	2.5 (1.15)
LD_{50} - dose causing death in 50% rats calculated from probit plot: mg.kg^{-1}	43.6	>84*	30.3
therapeutic index [LD50 ÷ AD50]	14.8	>30*	6.5

NB * and * - an accurate LD_{50} not possible for Phaxan$_{CD}$™

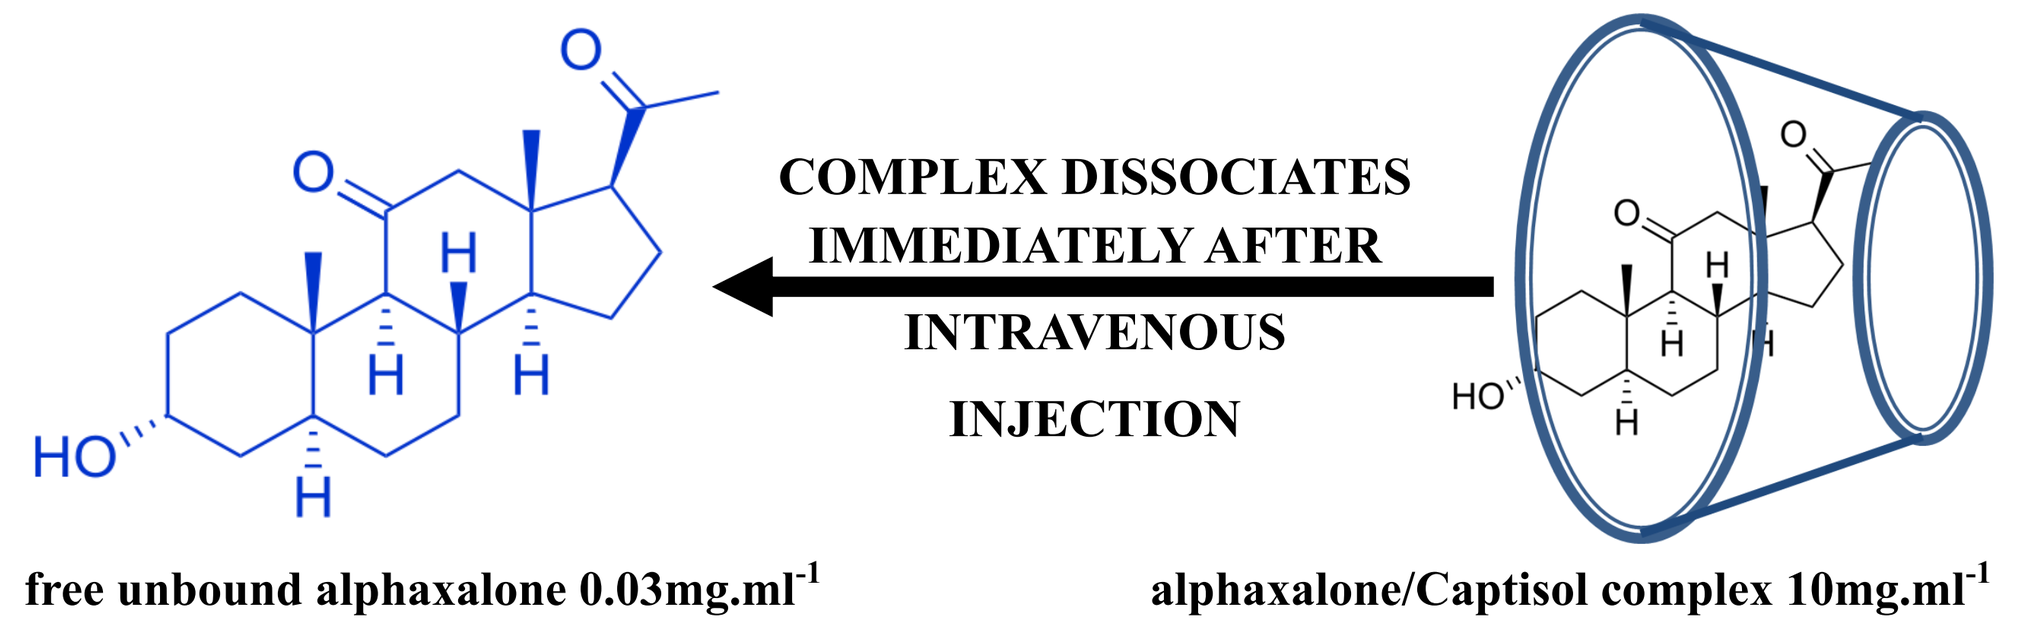
COMPLEX DISSOCIATES IMMEDIATELY AFTER INTRAVENOUS INJECTION
free unbound alphaxalone 0.03mg.ml^{-1} alphaxalone/Captisol complex 10mg.ml^{-1}

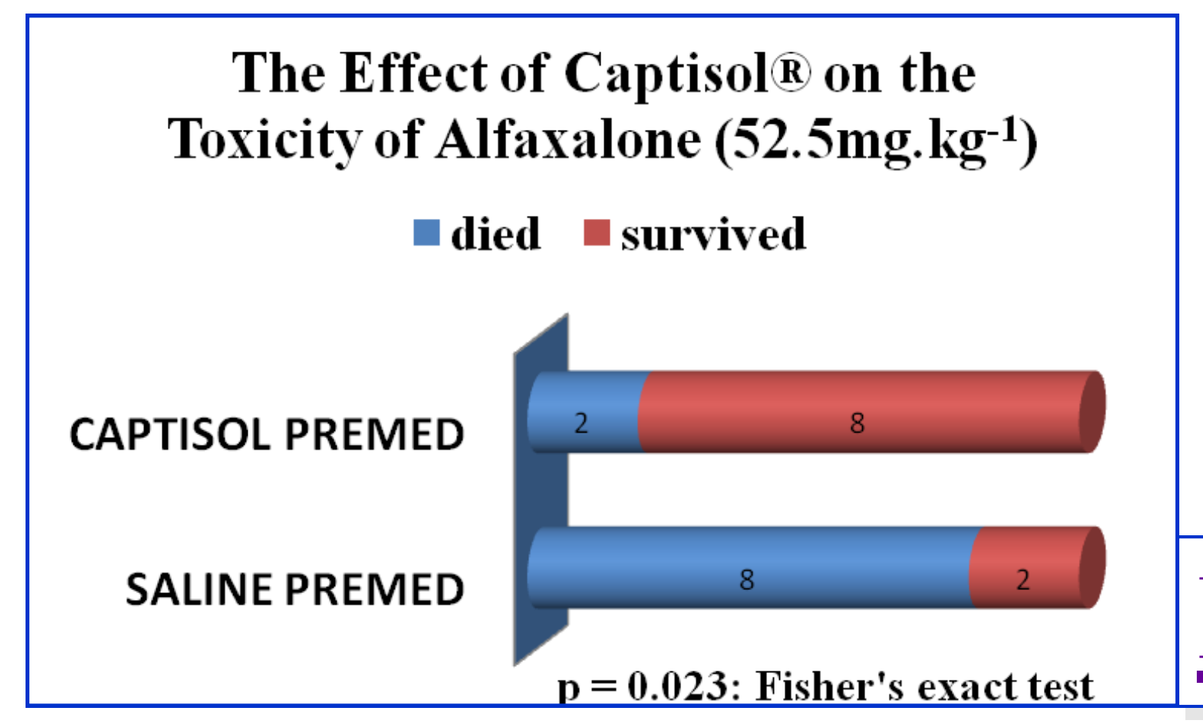
The Effect of Captisol® on the Toxicity of Alfaxalone (52.5mg.kg-1)
died
survived
CAPTISOL PREMED
SALINE PREMED
p = 0.023: Fisher's exact test
H

CONCLUSIONS

- **Phaxan$_{CD}$™** causes anesthesia with fast onset, and offset timing equal with propofol and Althesin®, the former commercial human anesthetic formulation of alphaxalone
- **Phaxan$_{CD}$™** is twice as potent as propofol
- **Phaxan$_{CD}$™** is a clear water-soluble preparation using an FDA-approved excipient to achieve water solubility—Captisol®. This avoids the only impediment to alphaxalone being used clinically for human anesthesia in todays operating rooms, i.e., hypersensitivity reactions to CremophorEL. This advance will allow the reintroduction of this useful agent into human anesthetic practice
- **Phaxan$_{CD}$™** is filterable with none of the infection, manufacturing and lipid toxicity issues associated with propofol lipid formulations
- **Phaxan$_{CD}$™** is less toxic than the propofol lipid formulation and also the alphaxalone formulation in CremophorEL (Althesin®)
- The Captisol® excipient in **Phaxan$_{CD}$™** is responsible for this by controlling the release of the alphaxalone from the complex

MATERIALS AND METHODS

A solution of alphaxalone [10mg.ml^{-1} ;Phaxan$_{CD}$™ (PHAX)] was prepared by dissolving alphaxalone 300mg [0.9 mmoles] in 30ml saline 0.9%, using Captisol® [7-sulfobutyl ether beta cyclodextrin] 3889mg [1.8 mmoles]. An "Althesin®-like" solution of alphaxalone [ALTH] was prepared in 20% CremophorEL as described in the literature (2). A propofol lipid emulsion [10mg.ml^{-1} in 10% Intralipid] was obtained from our pharmacy. Male Wistar rats [wt 150-220g] were implanted with internal jugular vein catheters under halothane anaesthesia. The following series' of experiments were performed 24 hours after intravenous cannulation.

Series 1: anesthesia and recovery

Each rat received a 15second intravenous injection from a range of doses of either PHAX, ALTH or PROP [1.25, 2.5, 5, 10, & 15mg.kg^{-1}; groups of n = 10 rats at each dose]. Three more groups of 10 rats each were given 1.5ml.kg^{-1}vehicle only [13% Captisol®; 20% CremophorEL; 10% lipid emulsion]. The following were assessed at regular time intervals after the intravenous injection:

- righting reflex: 1 normal; 2 slow; 3 some attempt; 4 none – this was a measure of onset and duration of unconsciousness; score 4 = unconscious and anaesthetised
- tail pinch response: 1 normal; 2 weak; 3 just present; 4 none – this was a measure of onset and duration of surgical anaesthesia; score 4 = surgically anaesthetised
- time the rat was able to walk/run on a rotating cylinder [rotarod] measured in seconds: the normal run time is 120 seconds in non sedated rats. The time after the intravenous anesthetic injection to achieve this score was the time taken for full recovery from the sedating effects of the anesthetic injection.

Results from groups of ten rats treated with the same anaesthetic and dose were combined for statistical purposes. The mean rotarod run times for rats in each treatment group were calculated and plotted graphically to show time of onset and recovery. The proportions of rats in each group of 10 similarly treated animals that scored 4 were entered into probit regression analysis using SPSS Statistics 18 [probit plot]. The estimated doses that caused anaesthesia in 50% and 95% of subjects [AD_{50} and AD_{95} respectively] for unconsciousness [righting reflex measurements] and surgical anaesthesia [tail pinch responses] were calculated from these plots.

Series 2: lethality and therapeutic index

Each rat received a 15second intravenous injection from a range of doses of either PHAX, ALTH or PROP [range 3-26 times AD_{95} for loss of righting reflex calculated for each agent from series 1 experiments; groups of n = 10 rats at each dose]. Three more groups of 10 rats each were given vehicle only [9.0 ml.kg^{-1} 13% Captisol®; 4.6 ml.kg^{-1}20% CremophorEL; 2,9 ml.kg^{-1}10% lipid emulsion]. The proportions of rats in each group of 10 similarly treated animals that died were entered into probit regression analysis using SPSS Statistics 18 [probit plot]. The estimated doses that caused death in 50% of subjects [LD_{50}] for each treatment were calculated from these plots. The therapeutic index for each agent was calculated by LD_{50}/AD_{50}; the higher the number meaning a safer agent.

Series 3: cardiovascular effects

Six groups of rats [n=5 each] were given AD_{95} doses of the anesthetics [PHAX 4.26mg.kg^{-1}; ALTH 4.39mg.kg^{-1}; PROP 5.4mg.kg^{-1}] or equivalent doses of vehicle only [Captisol® 0.43ml.kg^{-1}; CremophorEL 0.44ml.kg^{-1}; Intralipid 0.54ml.kg^{-1}]. Blood pressure and heart rate were measured every 5 min for 10 min before and 20min after these injections using a non invasive tail cuff blood pressure recorder with piezo-ceramic pulse detection. The readings taken at each time point from all rats in each group were averaged, plotted on time response curves and also compared for differences between treatments using repeated measures ANOVA.

Series 4: effect of Captisol® on alphaxalone toxicity

Two groups of rats [n=10 each] were given an intravenous premed injection 5 minutes before ALTH 52.5mg.kg^{-1} [5.25 mls.kg^{-1}; 12 times the AD_{95} dose which was expected to cause a high (>70%) mortality rate]. In one group the premed was 5.25ml.kg^{-1} saline 0.9% and in the other group of 10 rats the premed was 5.25ml.kg^{-1} Captisol® 13% solution. The proportion of survivors in each group was compared using Fishers exact test.

RESULTS

Alphaxalone [10mg.ml^{-1}] dissolved readily in 13% Captisol®-saline solution. The resultant solution was colourless and completely clear. It did not cause stiction when used in plastic syringes.

Series 1: anesthesia and recovery

Intravenous PHAX caused immediate dose-related sedation and anesthesia accompanied by no abnormal movements. The graphs D-F show the probit plots for the righting reflex anesthetic end point. Graphs A-C show the mean walking time on the rotarod for groups of 10 rats at each dose of the three anesthetic preparations. Table 1 summarises the results taken from these graphical plots. PHAX is equipotent with Althesin® in causing unconsciousness and surgical anaesthesia and both are more potent than PROP in this respect. Recovery from unconsciousness caused by PHAX is just as fast as with PROP. Control experiments revealed that the vehicles [20% CremophorEL, 10% lipid emulsion and 13% Captisol®] had no sedating or anaesthetic effects when given alone.

Series 2: lethality and therapeutic index

Doses of PROP greater than 30mg.kg^{-1} caused death in all rats. At doses of alphaxalone between 50 and 60mg/kg all the rats in the ALTH groups died, whereas none died in the PHAX groups that received the same doses of alphaxalone. The lethality for PHAX reached a ceiling at 20%; no more than 20% rats died even when the dose of alphaxalone administered as PHAX was increased. See probit plots for lethality D-F. The probit plots for ALTH and PROP were used to calculate the LD_{50} and LD_{95} values - see Table 1. A dose of 52mg.kg^{-1} alphaxalone as ALTH caused all 10 rats in that group to die but 64mg.kg^{-1} alphaxalone caused no deaths in the 10 rats which received that dose of PHAX. When the dose of alphaxalone as PHAX was increased to 71, 78 and then 84 mg.kg^{-1} only 2 of the 10 rats died in each group. Thus it was not possible to find the LD_{50} of alphaxalone as PHAX so a minimum estimated value is shown in Table 1 as >84mg.kg^{-1} [i.e., more than double the LD_{50} value for ALTH]. These results make the therapeutic index [ratio of LD_{50}:AD_{50}] to be 14.8 for ALTH and >30 for PHAX. Both of these contrast with the much lower therapeutic index for PROP in these experiments [6.5; Table 1]. No excipient caused death in any rat indicating that the difference in the safety/lethality of the two formulations of alphaxalone was not due to toxicity of the excipients.

Series 3: cardiovascular effects

Figure G shows the effect of equipotent doses of the three anaesthetics on systolic blood pressure. PHAX caused significantly smaller and shorter lasting falls in systolic blood pressure than PROP.

Series 4: effect of Captisol® on alphaxalone toxicity

Figure H shows the results of experiments in which Captisol® or saline were given prior to a toxic dose of alphaxalone as ALTH. The Captisol® caused a significant reduction in lethality suggesting that the Captisol® in PHAX is responsible for the higher therapeutic index of PHAX compared with ALTH. The control experiments with CremophorEL and Captisol® given alone showed no deaths.

A

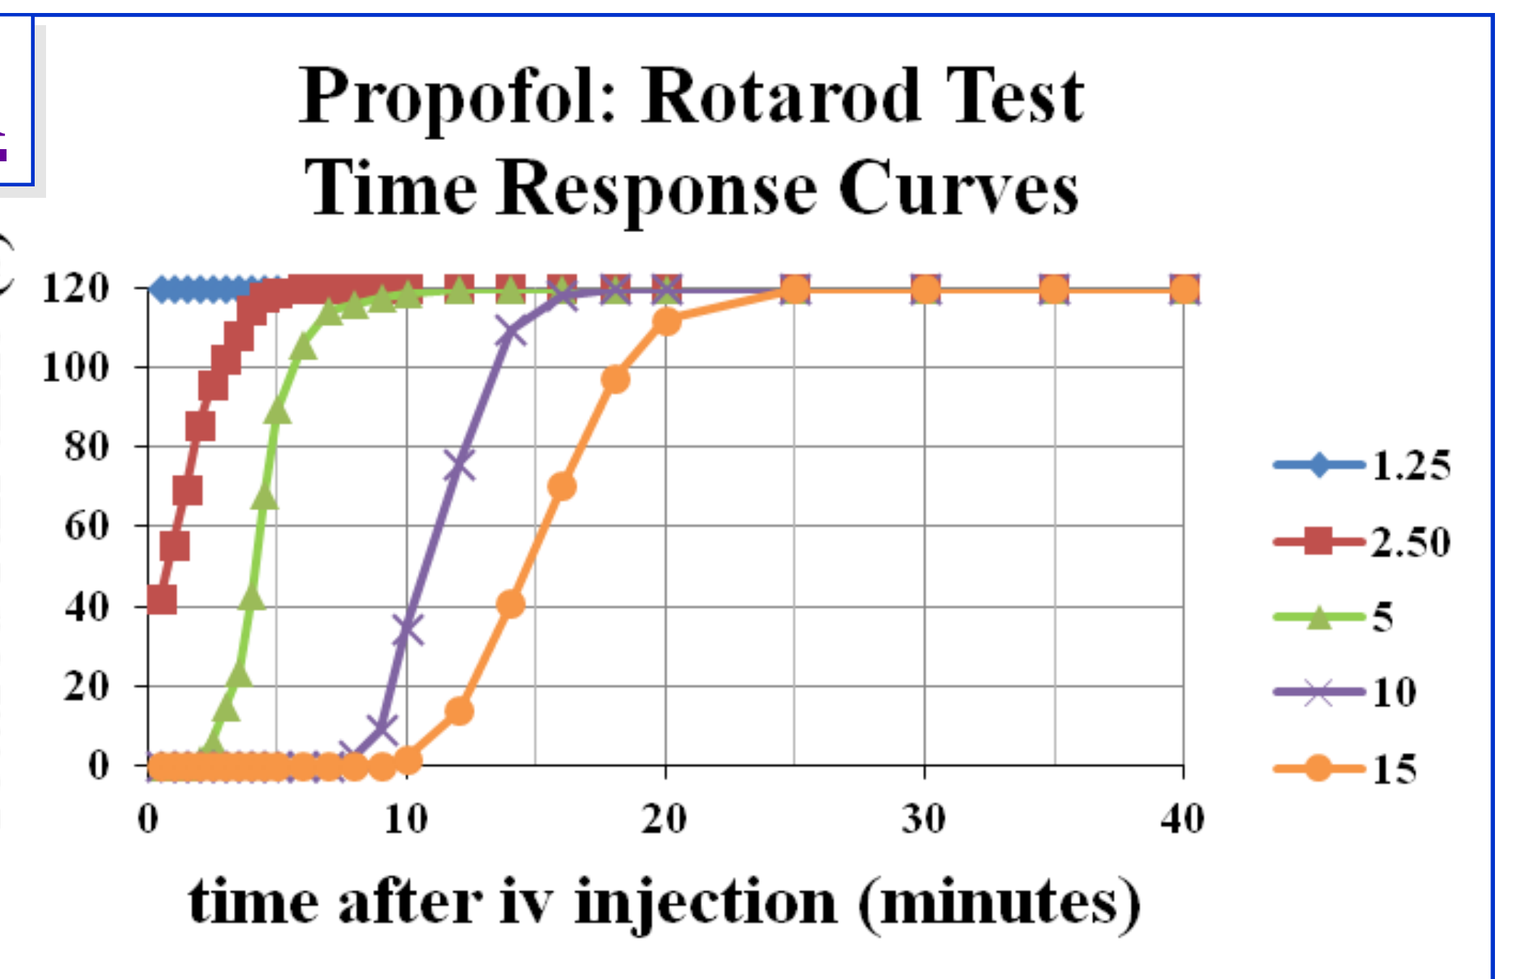
Propofol: Rotarod Test
Time Response Curves
rotarod run time (s)
time after iv injection (minutes)
1.25
2.50
5
10
15

D

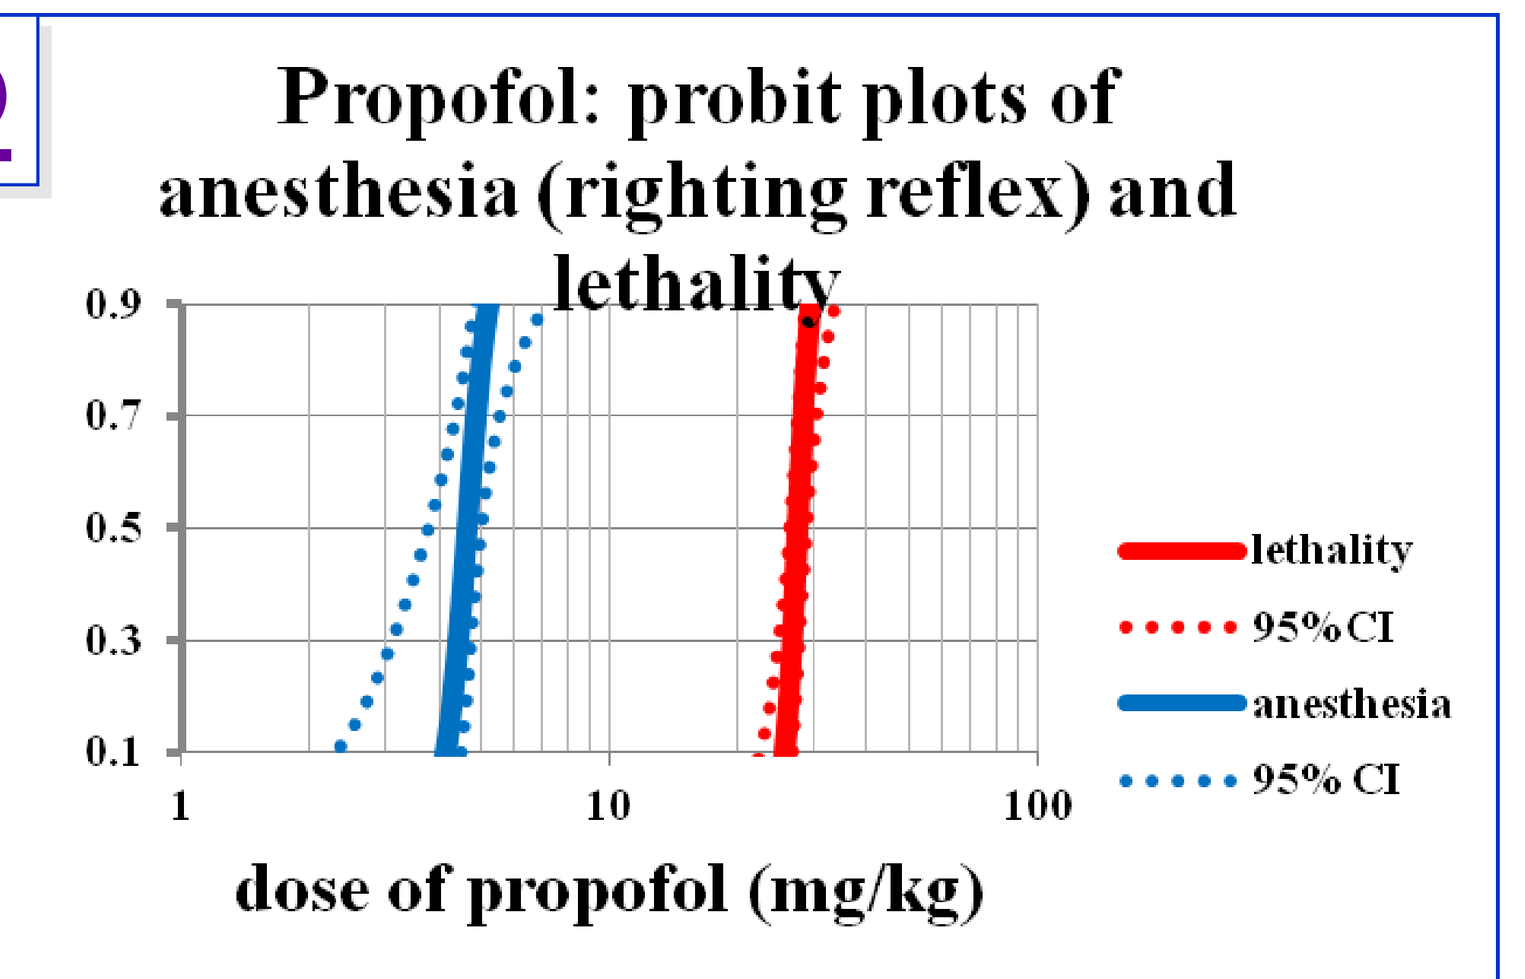
Propofol: probit plots of anesthesia (righting reflex) and lethality
probit value
dose of propofol (mg/kg)
lethality
95%CI
anesthesia
95% CI

B

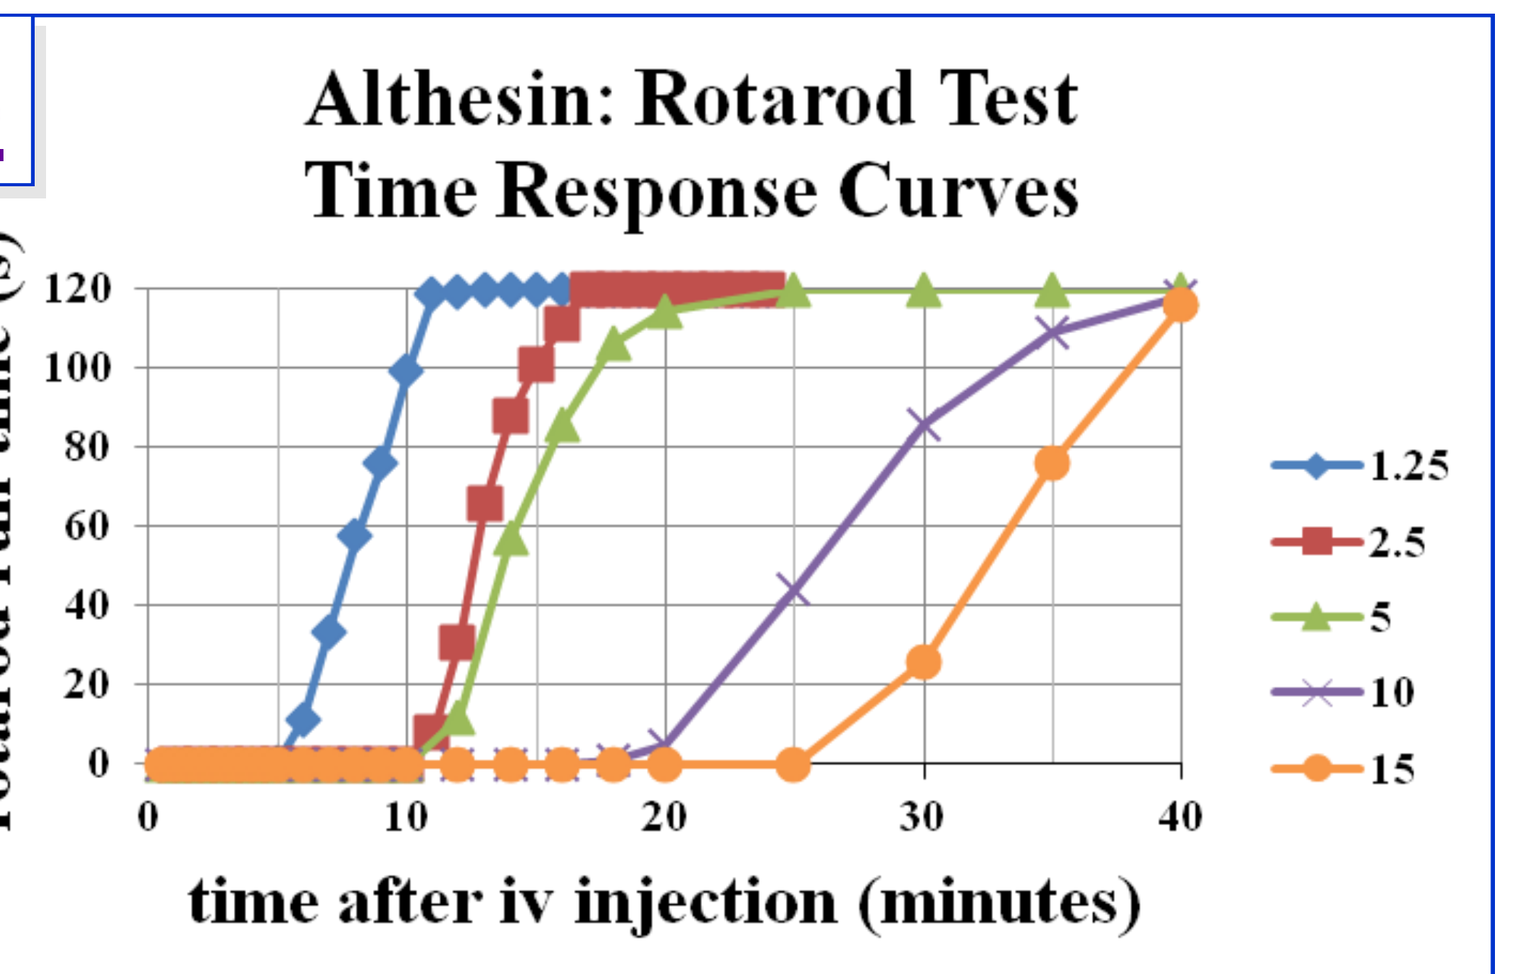
Althesin: Rotarod Test
Time Response Curves
rotarod run time (s)
time after iv injection (minutes)
1.25
2.5
5
10
15

E

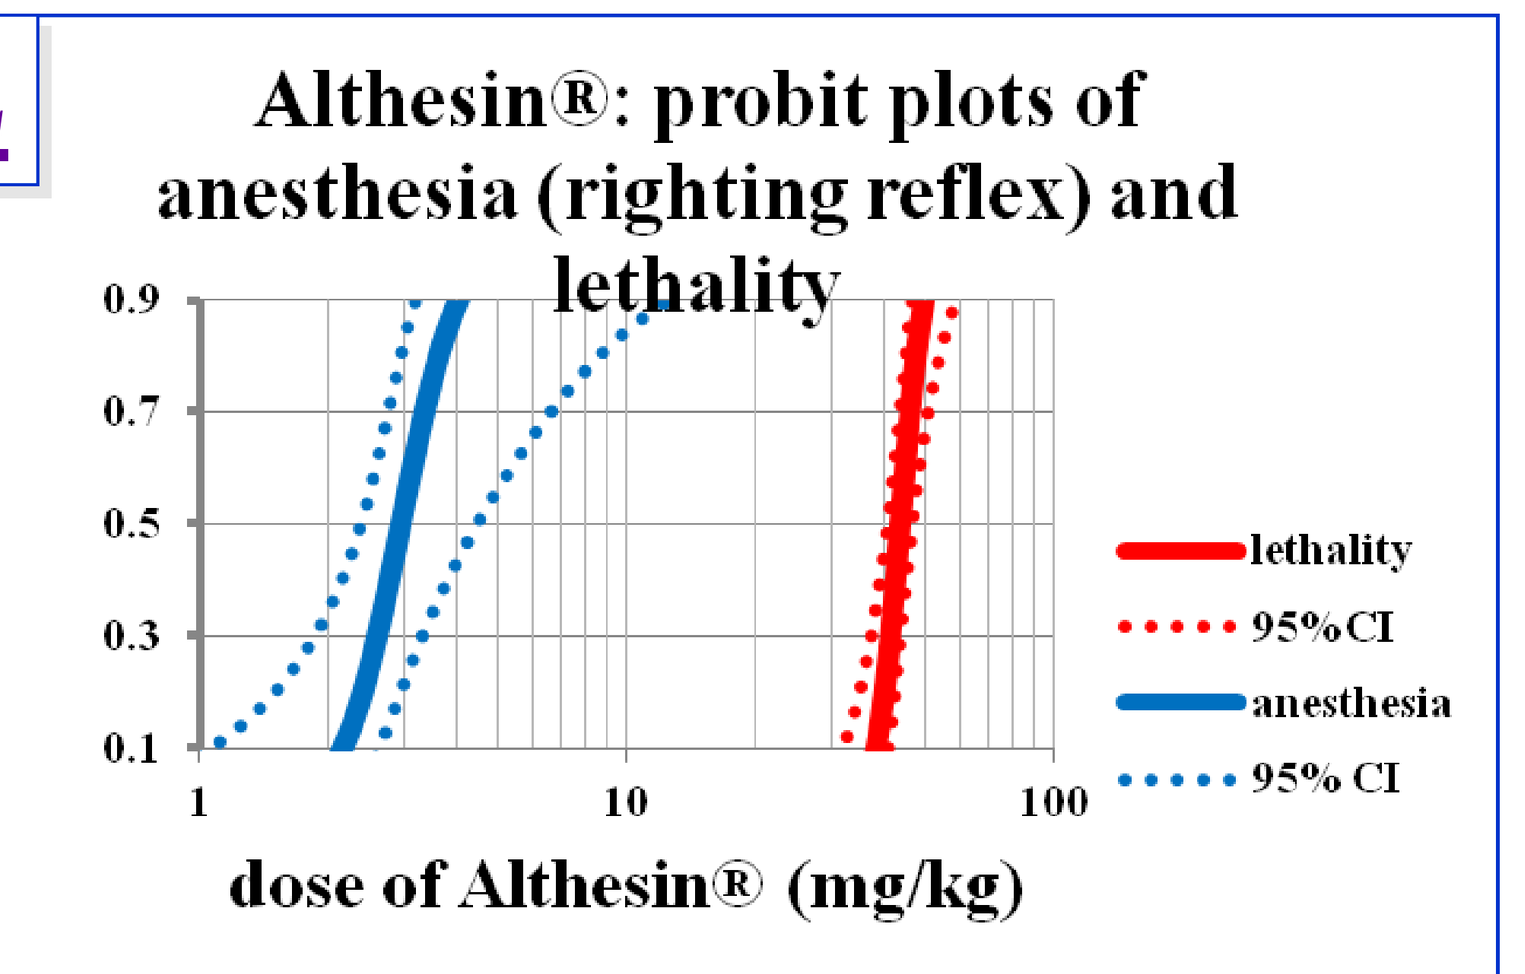
Althesin®: probit plots of anesthesia (righting reflex) and lethality
probit value
dose of Althesin® (mg/kg)
lethality
95%CI
anesthesia
95% CI

C

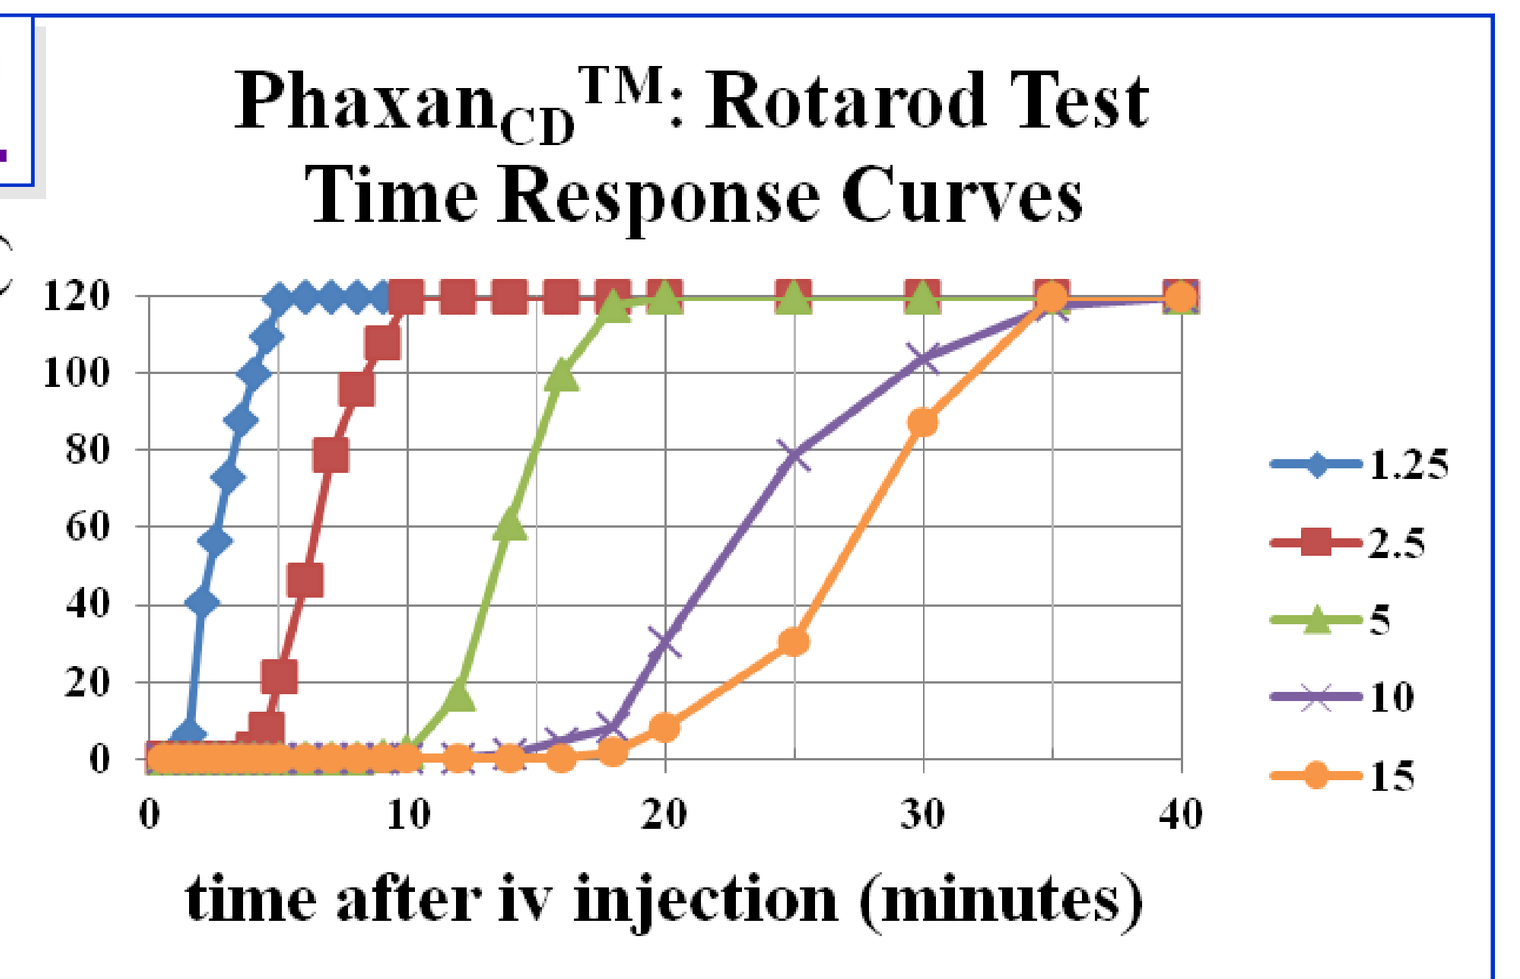
PhaxanCD™: Rotarod Test
Time Response Curves
rotarod run time (s)
time after iv injection (minutes)
1.25
2.5
5
10
15

F

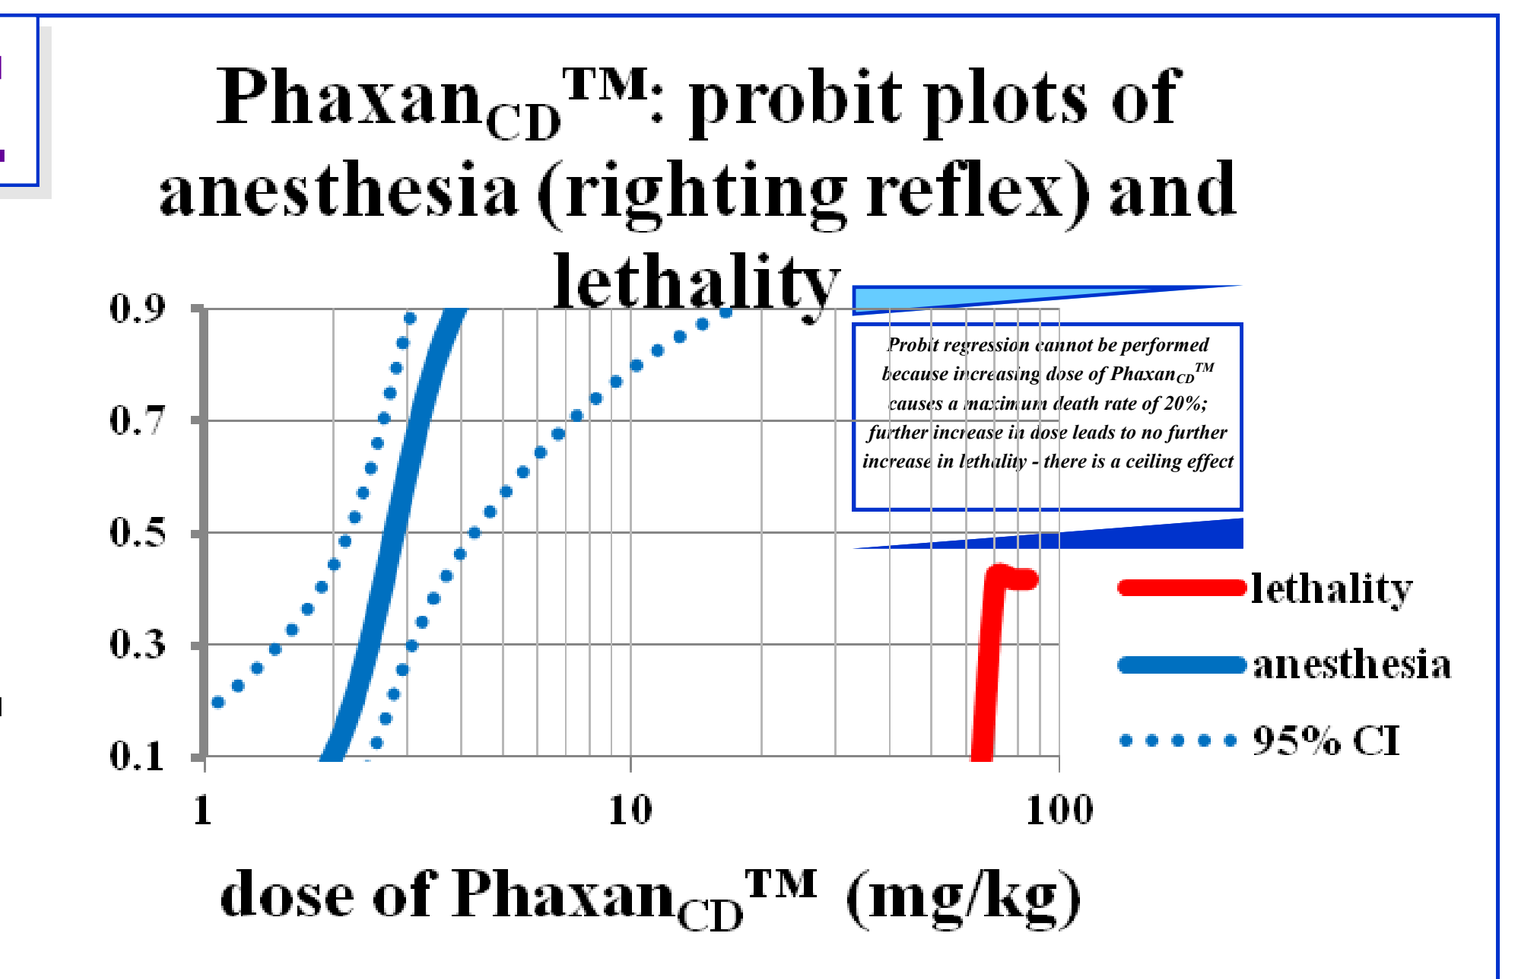
PhaxanCD™: probit plots of anesthesia (righting reflex) and lethality
probit value
dose of PhaxanCD™ (mg/kg)
lethality
anesthesia
95% CI

G

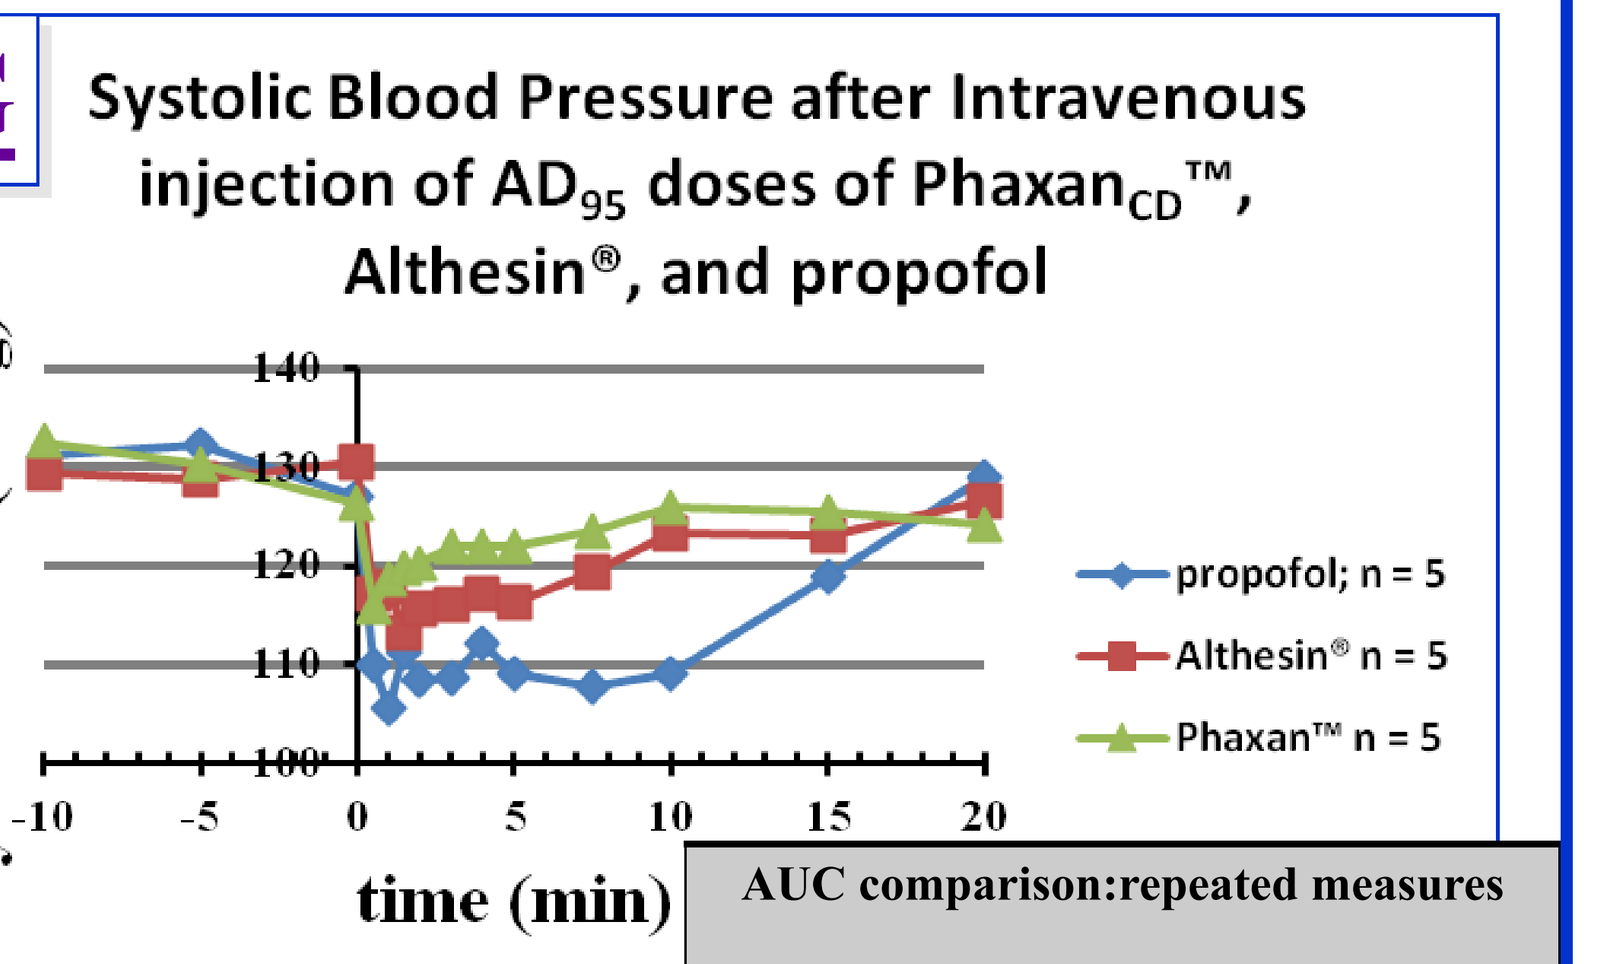
Systolic Blood Pressure after Intravenous injection of AD95 doses of PhaxanCD™, Althesin®, and propofol
systolic BP (mHg)
time (min)
propofol; n = 5
Althesin® n = 5
Phaxan™ n = 5

AUC comparison:repeated measures			
p value: Tukey Kramer post hoc test	propofol v Althesin® >0.05	propofol v Phaxan$_{CD}$™ <0.01	Althesin® v Phaxan$_{CD}$™ >0.05

The authors wish to thank and acknowledge Cydex Pharmaceuticals Inc, (a company owned by Ligand Pharmaceuticals, La Jolla, CA) for supplies of Captisol®

Captisol® is a patent protected, modified cyclodextrin: 7-sulfobutyl ether beta cyclodextrin
Phaxan is patent protected and a registered trade mark for alphaxalone dissolved in 13% Captisol®

MONASH INSTITUTE OF MEDICAL RESEARCH
MONASH University Pharmacy and Pharmaceutical Sciences